Efficacy of Zanubrutinib Versus Acalabrutinib in the Treatment of Relapsed or Refractory Chronic Lymphocytic Leukemia (R/R CLL): A Matching-Adjusted Indirect Comparison (MAIC)

Mazyar Shadman[1], Jennifer R Brown[2], Rhys Williams[3], Leyla Mohseninejad[4], Keri Yang[3], Pal Rakonczai[5], Nicole Lamanna[6], Sheng Xu[7], Aileen Cleary Cohen[3], Susan M. O'Brien[8], Alessandra Tedeschi[9], and Constantine S. Tam[10]

[1]Fred Hutchinson Cancer Center and University of Washington, Seattle, WA, USA; [2]Chronic Lymphocytic Leukemia Center, Division of Medical Oncology, Dana–Farber Cancer Institute and Harvard Medical School, Boston, MA, USA; [3]BeiGene USA, Inc, San Mateo, CA, USA; [4]BeiGene Netherlands B.V., Schiphol, Netherlands; [5]Evidera, Budapest, Hungary; [6]Herbert Irving Comprehensive Cancer Center, Columbia University Medical Center, New York, NY, USA; [7]BeiGene (Shanghai) Co., Ltd., Shanghai, China; [8]University of California, Irvine, CA, USA; [9]ASST Grande Ospedale Metropolitano Niguarda, Milan, Italy; [10]Alfred Hospital and Monash University, Melbourne, VIC, Australia

BACKGROUND AND OBJECTIVE

- Zanubrutinib, a next-generation covalent Bruton tyrosine kinase inhibitor (BTKi), is the only BTKi that demonstrated progression-free survival (PFS) superiority vs ibrutinib (first-generation BTKi) in relapsed or refractory (R/R) chronic lymphocytic leukemia (CLL) in the ALPINE trial.[1]
- Acalabrutinib, a second-generation BTKi, showed improved PFS vs rituximab-idelalisib/bendamustine in R/R CLL in the ASCEND trial,[2,3] but PFS noninferiority vs ibrutinib in patients with R/R CLL with chromosome 17p or 11q deletions in the ELEVATE-RR trial.[4]
- As no head-to-head clinical trial of zanubrutinib and acalabrutinib in R/R CLL exists, an indirect treatment comparison was performed to evaluate the relative efficacy of these two treatments.
- The objective of this study was to compare the efficacy of zanubrutinib in ALPINE and acalabrutinib in ASCEND using matching-adjusted indirect comparison (MAIC) methodology.

METHODOLOGY

- Individual patient-level data (IPD) from ALPINE was matched against the aggregate data from ASCEND.[1-3]
- An unanchored MAIC was used due to the lack of a common comparator arm between the ALPINE and ASCEND trials.
- Given the timing of the study in relation to the COVID-19 pandemic for ASCEND vs ALPINE, adjustments on ALPINE were made for the impact of COVID-19.
- Population adjustment in the base case analysis considered all variables identified as prognostic factors or predictors of treatment effect **(Fig. 1**; **Table 1)**.
- Pseudo IPD for PFS and overall survival (OS) in the acalabrutinib arm of ASCEND were reconstructed from the digitized Kaplan-Meier curves reported in the ASCEND publication using the algorithm by Guyot et al.[5]
- A weighted Cox proportional hazard model was used to compare investigator-assessed PFS (PFS-INV) and OS and a weighted logistic regression model to compare complete response (CR).

Figure 1. Details of the overall methodology

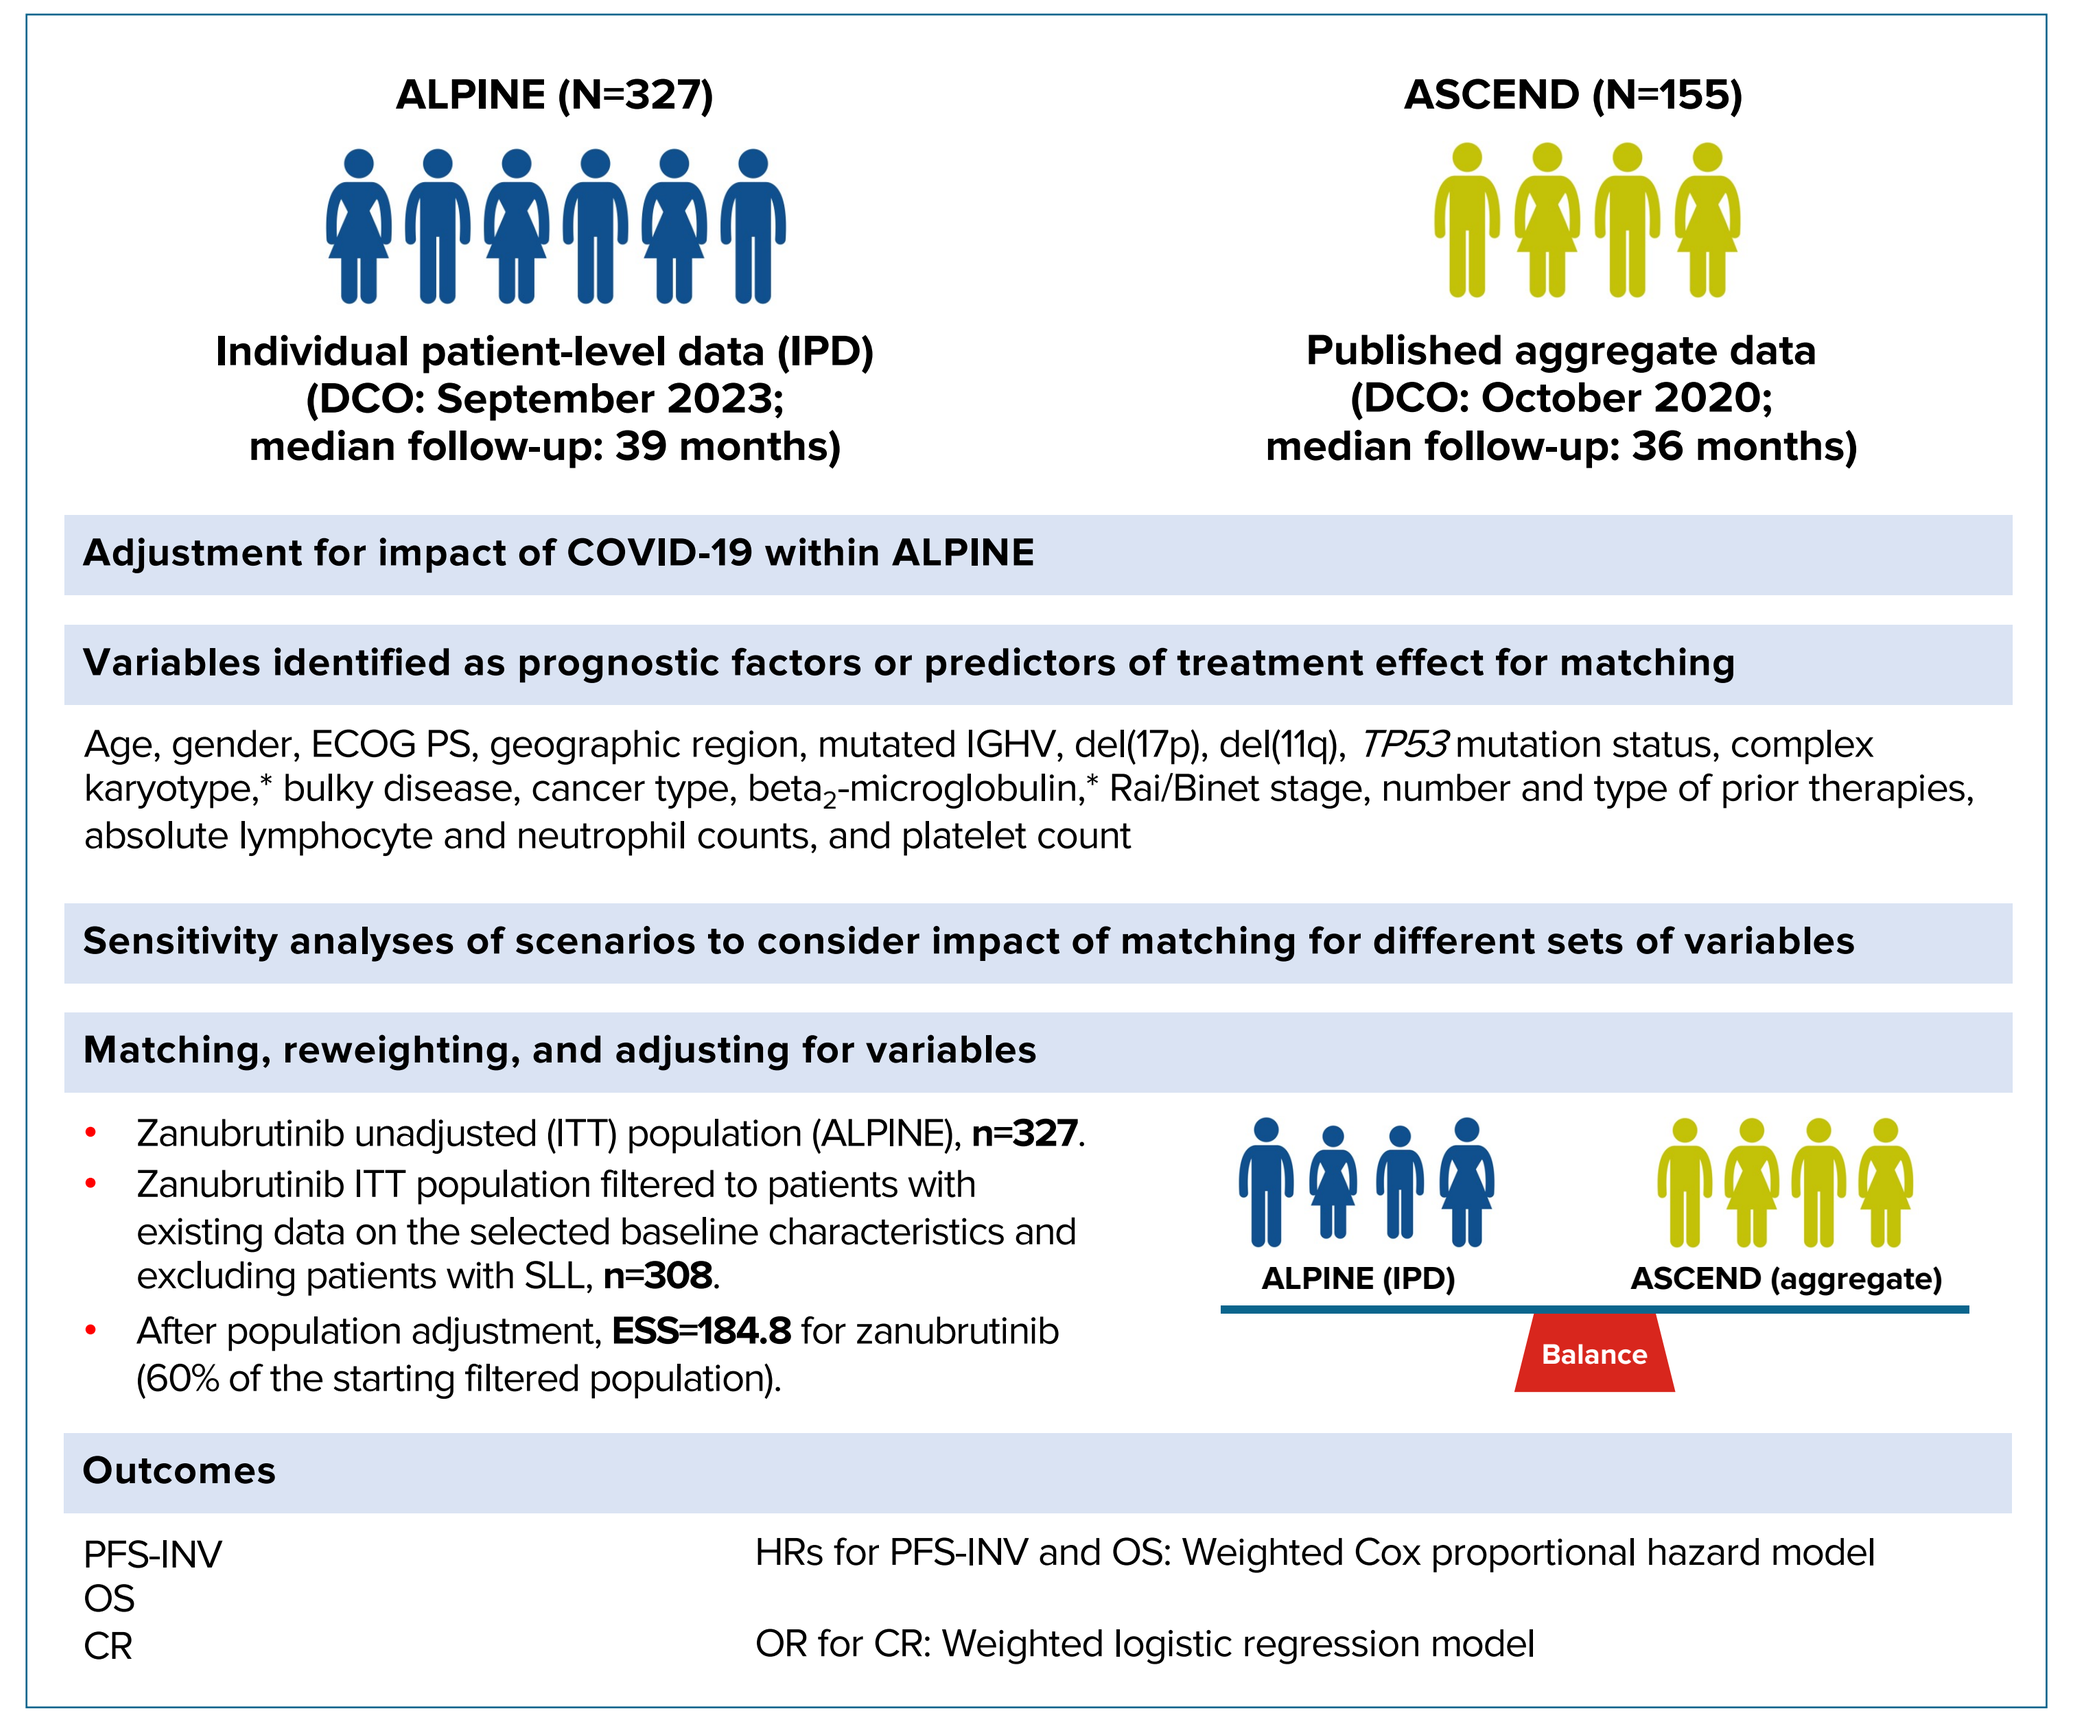
ALPINE (N=327)
Individual patient-level data (IPD)
(DCO: September 2023;
median follow-up: 39 months)
ASCEND (N=155)
Published aggregate data
(DCO: October 2020;
median follow-up: 36 months)
Adjustment for impact of COVID-19 within ALPINE
Variables identified as prognostic factors or predictors of treatment effect for matching
Age, gender, ECOG PS, geographic region, mutated IGHV, del(17p), del(11q), TP53 mutation status, complex karyotype,* bulky disease, cancer type, beta2-microglobulin,* Rai/Binet stage, number and type of prior therapies, absolute lymphocyte and neutrophil counts, and platelet count
Sensitivity analyses of scenarios to consider impact of matching for different sets of variables
Matching, reweighting, and adjusting for variables
Zanubrutinib unadjusted (ITT) population (ALPINE), n=327.
Zanubrutinib ITT population filtered to patients with existing data on the selected baseline characteristics and excluding patients with SLL, n=308.
After population adjustment, ESS=184.8 for zanubrutinib (60% of the starting filtered population).
ALPINE (IPD)
ASCEND (aggregate)
Balance
Outcomes
PFS-INV
OS
CR
HRs for PFS-INV and OS: Weighted Cox proportional hazard model
OR for CR: Weighted logistic regression model

*Covariates not matched in the base case.
CR, complete response; DCO, data cut-off; del(11q), chromosome 11q deletion; del(17p), chromosome 17p deletion; ECOG PS, Eastern Cooperative Oncology Group performance status; ESS, effective sample size; HR, hazard ratio; IGHV, immunoglobulin heavy chain variable; IPD, individual patient-level data; ITT, intent to treat; OR, odds ratio; OS, overall survival; PFS-INV, investigator-assessed progression-free survival; SLL, small lymphocytic lymphoma.

METHODOLOGY

Table 1. Covariates matched in the base case and sensitivity analyses

Covariates	Main analysis		Sensitivity analyses					
	Unadjusted population	Base case adjusted population	S1	S2	S3	S4	S5	S6
Age ≥75, %		✓	✓	✓	✓	✓	✓	✓
Male, %		✓	✓	✓	✓	✓	✓	✓
ECOG PS score=0 (vs. ≥1), %		✓	✓		✓	✓	✓	✓
Geographic region								
United States and Canada, %		✓	✓	✓	✓	✓	✓	✓
Australia and New Zealand, %		✓	✓	✓	✓	✓	✓	✓
Asia, %		✓	✓	✓	✓	✓	✓	✓
Europe, %		✓	✓	✓	✓	✓	✓	✓
Genomic status								
Mutated IGHV, %		✓	✓	✓	✓	✓	✓	✓
Del(17p), %		✓	✓	✓	✓	✓	✓	✓
Del(11q), %		✓	✓	✓	✓	✓	✓	✓
TP53 mutation, %		✓	✓	✓	✓	✓	✓	✓
Complex karyotype ≥3, %*								✓
Bulky disease, LDi in cm, ≥5, %		✓	✓	✓	✓	✓	✓	✓
Cancer type, CLL, %		✓	✓	✓	✓	✓	✓	✓
Beta$_2$-microglobulin >3.5 mg/L, %*								✓
Rai stage 0-II or Binet A/B, %		✓	✓	✓	✓	✓	✓	✓
Number of prior therapies								
2, %		✓	✓	✓	✓			✓
3, %		✓	✓	✓	✓			✓
≥4, %		✓	✓	✓	✓		✓	✓
Prior therapy								
Anti-CD20 antibody, %		✓	✓			✓	✓	✓
Alkylators other than bendamustine, %		✓	✓			✓	✓	✓
Bendamustine, %		✓	✓			✓	✓	✓
Purine analog, %		✓	✓			✓	✓	✓
Absolute lymphocyte count, 10^9 cells/L, median		✓				✓	✓	✓
Absolute neutrophil count, 10^9 cells/L, median		✓				✓	✓	✓
Platelet count, 10^9 cells/L, median		✓				✓	✓	✓

*Covariates not matched in the base case.
Del (11q), chromosome 11q deletion; del (17p), chromosome 17p deletion; ECOG PS, Eastern Cooperative Oncology Group performance status; IGHV, immunoglobulin heavy chain variable; LDi, longest diameter.

RESULTS

Table 2. Baseline characteristics of the zanubrutinib and acalabrutinib populations

Covariates	Acalabrutinib ASCEND (N=155)	Zanubrutinib ALPINE (N=327)	Zanubrutinib ALPINE post-matching (ESS=184.8)
Age ≥75, %	21.9	22.6	21.9
Male, %	69.7	65.1	69.7
ECOG PS score=0 (vs. ≥1), %	37.4	39.9	37.4
Geographic region			
United States and Canada, %	**5.2**	**15.9**	5.2
Australia and New Zealand, %	5.8	8.6	5.8
Asia, %	**4.5**	**15.0**	4.5
Europe, %	**84.5**	**60.6**	84.5
Genomic status			
Mutated IGHV, %	**16.2**	**25.0**	16.2
Del(17p), %	17.4	13.8	17.4
Del(11q), %	25.2	27.8	25.2
***TP53* mutation, %**	**25.2**	**15.3**	25.2
Complex karyotype ≥3, %*	32.4	26.8	28.6
Bulky disease, LDi in cm, ≥5, %	49.0	44.3	49.0
Cancer type, CLL, %	100	96	100
Beta$_2$-microglobulin >3.5 mg/L, %*	77.4	62.6	62.8
Rai stage 0-II or Binet A/B, %	58.1	58.0	58.1
Number of prior therapies			
2, %	25.8	26.3	25.8
3, %	11.0	7.6	11.0
≥4, %	10.3	7.3	10.3
Prior therapy			
Anti-CD20 antibody, %	83.9	83.8	83.9
Alkylators other than bendamustine, %	85.8	83.8	85.8
Bendamustine, %	30.3	25.7	30.3
Purine analog, %	**70.3**	**54.4**	70.3
Absolute lymphocyte count, 10^9 cells/L, median	48.9	36.0	49
Absolute neutrophil count, 10^9 cells/L, median	3.8	4.0	4
Platelet count, 10^9 cells/L, median	119.5	126.0	119.0

Bold values imply a statistically significant difference between zanubrutinib and acalabrutinib pre- matching.
*Covariates not matched in the base case.
del(11q), chromosome 11q deletion; del(17p), chromosome 17p deletion; ECOG PS, Eastern Cooperative Oncology Group performance status; ESS, effective sample size; IGHV, Immunoglobulin heavy chain variable; LDi, longest diameter.

Efficacy outcomes

- PFS-INV was significantly improved for zanubrutinib post-matching (**Fig 2A**); OS was potentially improved for zanubrutinib post-matching **(Fig 2B)**.
- CR favored zanubrutinib in the unadjusted and base case adjusted populations (**Table 3**).
- Results for the sensitivity analyses were consistent with the base case (**Table 3**).

Figure 2. (A) PFS-INV and (B) OS for zanubrutinib ITT and post-matching, and acalabrutinib

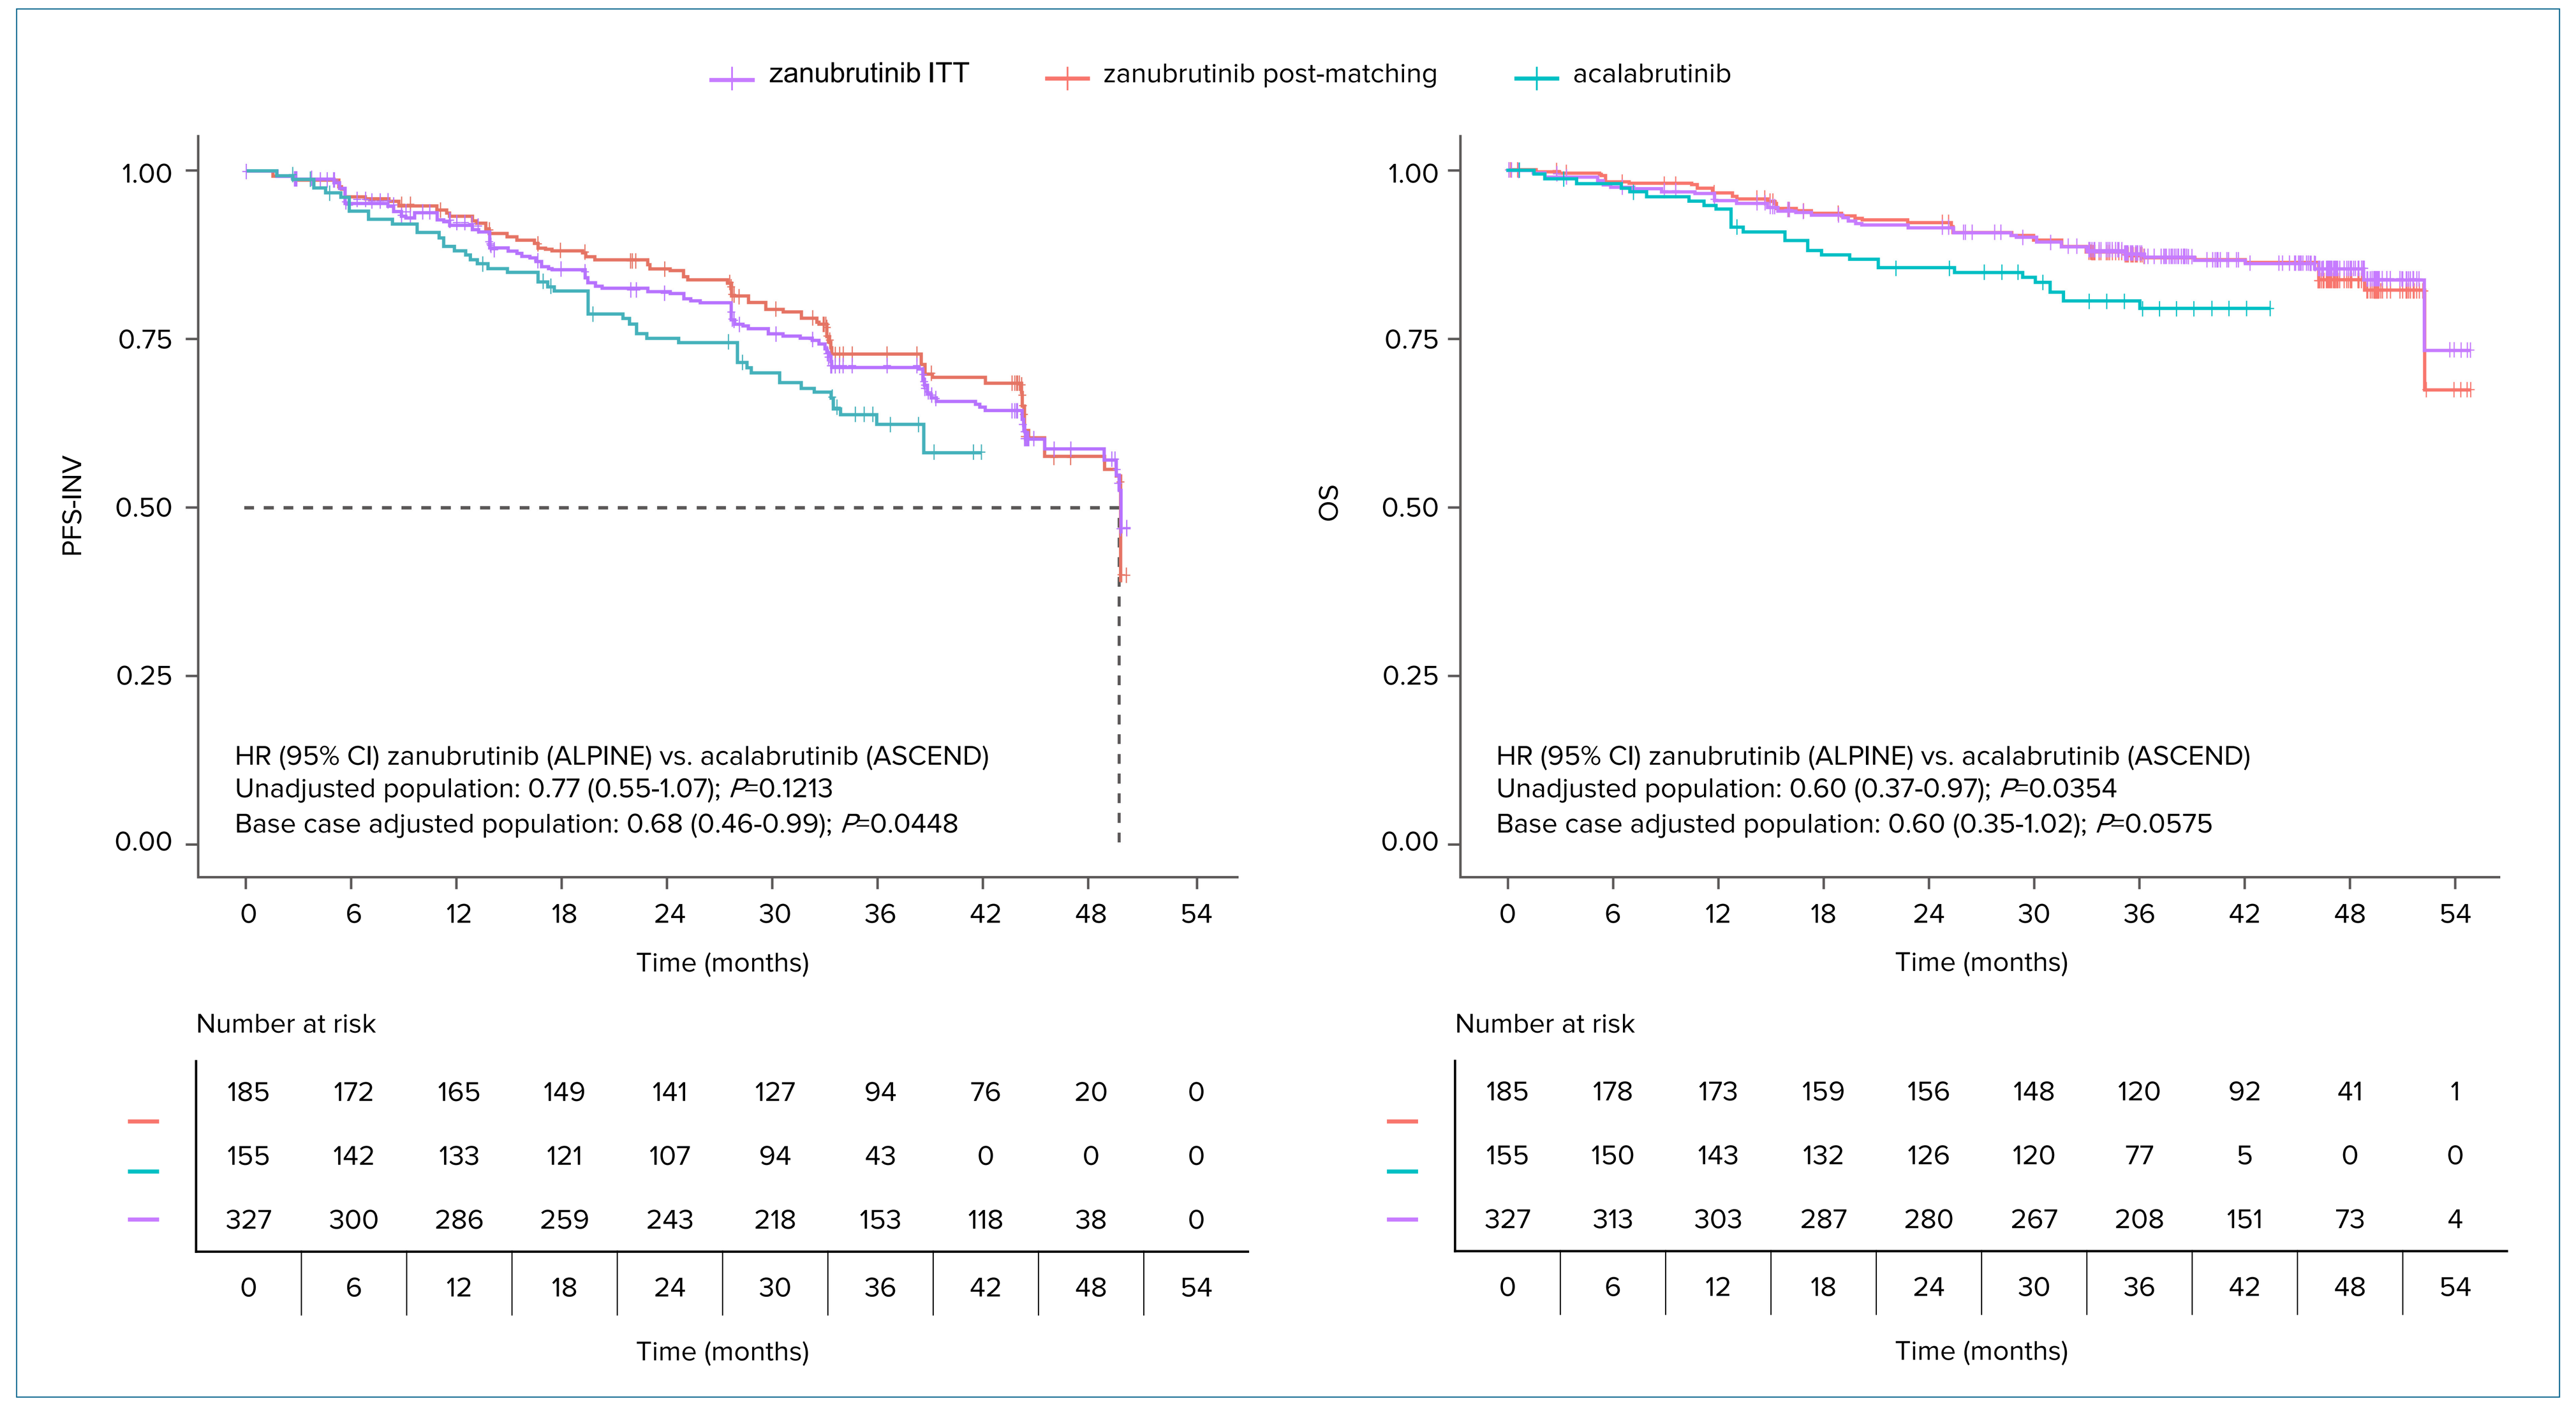
zanubrutinib ITT
zanubrutinib post-matching
acalabrutinib
PFS-INV
1.00
0.75
0.50
0.25
0.00
HR (95% CI) zanubrutinib (ALPINE) vs. acalabrutinib (ASCEND)
Unadjusted population: 0.77 (0.55-1.07); P=0.1213
Base case adjusted population: 0.68 (0.46-0.99); P=0.0448
0 6 12 18 24 30 36 42 48 54
Time (months)
Number at risk
185 172 165 149 141 127 94 76 20 0
155 142 133 121 107 94 43 0 0 0
327 300 286 259 243 218 153 118 38 0
0 6 12 18 24 30 36 42 48 54
Time (months)
OS
1.00
0.75
0.50
0.25
0.00
HR (95% CI) zanubrutinib (ALPINE) vs. acalabrutinib (ASCEND)
Unadjusted population: 0.60 (0.37-0.97); P=0.0354
Base case adjusted population: 0.60 (0.35-1.02); P=0.0575
0 6 12 18 24 30 36 42 48 54
Time (months)
Number at risk
185 178 173 159 156 148 120 92 41 1
155 150 143 132 126 120 77 5 0 0
327 313 303 287 280 267 208 151 73 4
0 6 12 18 24 30 36 42 48 54
Time (months)

CI, confidence interval; HR, hazard ratio; ITT, intent-to-treat; OS, overall survival; PFS-INV, investigator-assessed progression-free survival.

Table 3. Relative treatment effects for base case and sensitivity analyses

	Main analysis		Sensitivity analyses					
	Unadjusted population	Base case adjusted population	S1	S2	S3	S4	S5	S6
Sample size for ALPINE zanubrutinib	N=327	ESS=184.8	ESS=188.9	ESS=210.3	ESS=208.1	ESS=188.2	ESS=187.4	ESS=78.2
HR PFS-INV zanubrutinib vs. acalabrutinib (95% CI, *P* value)	0.77 (0.55-1.07, *P*=0.1213)	**0.68 (0.46-0.99, *P*=0.0448)**	**0.68 (0.47-1.00, *P*=0.0483)**	0.72 (0.5-1.04, *P*=0.0842)	0.73 (0.51-1.05, *P*=0.0921)	**0.67 (0.46-0.98, *P*=0.0410)**	**0.67 (0.46-0.98, *P*=0.0386)**	0.71 (0.43- 1.17, *P*=0.1822)
HR OS zanubrutinib vs. acalabrutinib (95% CI, *P* value)	**0.6 (0.37-0.97, *P*=0.0354)**	0.6 (0.35-1.02, *P*=0.0575)	**0.59 (0.35-1.00, *P*=0.0481)**	0.63 (0.38-1.04, *P*=0.0720)	0.66 (0.40-1.09, *P*=0.1030)	0.61 (0.36-1.03, *P*=0.0627)	0.61 (0.36-1.03, *P*=0.0667)	0.68 (0.33-1.39, *P*=0.2872)
OR CR zanubrutinib vs. acalabrutinib (95% CI, *P* value)	**2.88 (1.18-7.02, *P*=0.0198)**	**2.90 (1.13-7.43, *P*=0.0270)**	**2.88 (1.13-7.38, *P*=0.0273)**	**2.69 (1.06-6.85, *P*=0.0377)**	**2.78 (1.09-7.07, *P*=0.0316)**	**2.85 (1.11-7.31, *P*=0.0294)**	**2.80 (1.09-7.19, *P*=0.0326)**	**3.34 (1.15-9.71, *P*=0.0264)**

Bold values indicate P<0.05.
CI, confidence interval; CR, complete response; ESS, effective sample size; HR, hazard ratio; OR, odds ratio; OS, overall survival; PFS-INV, investigator-assessed progression-free survival.

CONCLUSIONS

- This comprehensive MAIC demonstrated a significant PFS and CR advantage, and potentially improved OS for zanubrutinib compared with acalabrutinib.
 - Results were robust across multiple sensitivity analyses.
- In a previous publication, Kittai et al. presented a MAIC to compare the efficacy and safety of zanubrutinib (ALPINE, aggregate) vs. acalabrutinib (ASCEND, IPD) in R/R CLL. Findings showed similar efficacy for zanubrutinib and acalabrutinib (PFS-INV) and different adverse event profiles.[7]
 - The efficacy results differ from those presented here because the analysis by Kittai et al. had several important limitations, including different follow-ups between ALPINE and ASCEND, lack of any adjustment for COVID-19, and incomplete matching variables (e.g., no granularity in geographic regions and number and types of prior therapies).
- While MAICs provide a scientific basis for evaluating hypotheses with regards to treatment efficacy across trials, the gold standard for evaluating evidence of relative efficacy remains randomized controlled trials.

LIMITATIONS

- There is a potential for bias resulting from the strong assumption that cross-trial differences can be entirely explained by variables selected for matching.
- Independent review committee-assessed PFS was not analyzed in the current MAIC due to unavailability of data in ASCEND and the latest ALPINE data cutoff.
- The study did not compare safety for zanubrutinib vs acalabrutinib, given different treatment exposure times across the two trials.
 - Safety of a drug is best evaluated via meta-analyses that use all available evidence across all indications.
 - A recent meta-analysis of 61 trials involving 6,959 patients who received ibrutinib, ibrutinib±anti-CD20 antibody, acalabrutinib, and zanubrutinib extensively analyzed the AE profiles of zanubrutinib and acalabrutinib across several indications and reported differences between the two treatments.[6]

DISCLOSURES

MS reports receiving funding from BeiGene paid to Fred Hutchinson Cancer Research Center and the University of Washington (Seattle, WA, USA), with regards to the submitted work; research funding from Pharmacyclics (Inst), Acerta Pharma (Inst), Merck (Inst), TG Therapeutics (Inst), BeiGene (Inst), Celgene (Inst), Genentech (Inst), MustangBio (Inst), AbbVie (Inst), Sunesis Pharmaceuticals (Inst), Bristol Myers Squibb/Celgene, Genmab (Inst), and Vincerx Pharma (Inst); and consulting or advisory roles with AbbVie, Genentech, AstraZeneca, Sound Biologics, Cellectar, Pharmacyclics, BeiGene, Bristol Myers Squibb/Celgene, MorphoSys, Innate Pharma, Kite, a Gilead Company, Adaptive Biotechnologies, Epizyme, Fate Therapeutics, Lilly, Regeneron, Adaptimmune, MustangBio, TG Therapeutics, and MEI Pharma, outside the submitted work.

REFERENCES

1. Brown JR, et al. *N Engl J Med.* 2023; 388: 319-332.
2. Ghia P, et al. *J Clin Oncol.* 2020; 38: 2849-2861.
3. Ghia P, et al. *Hemasphere.* 2022; 6(12):e801.
4. Byrd JC, et al. *J Clin Oncol.* 2021; 39: 3441–3452.
5. Guyot P, et al. *BMC Med Res Methodol.* 2012; 12: 9.
6. Hwang S, et al. *Hemasphere.* 2023;7(S3):1134.
7. Kittai AS, et al. *Am J Hematol.* 2023;98(12):E387-E390.

Presented at the 28th International Congress on Hematologic Malignancies, February 29 - March 3, 2024, Miami, FL